UNDERSTANDING HEMATOLOGICAL ADVERSE EVENT MANAGEMENT THROUGH HEALTH CARE RESOURCE UTILIZATION, COSTS, AND TREATMENT PATTERNS OF PATIENTS WITH EXTENSIVE-STAGE SMALL CELL LUNG CANCER TREATED IN THE COMMUNITY ONCOLOGY SETTING

Jerome Goldschmidt[1]; Alisha Monnette[2]; Ping Shi[2]; Divea Venkatasetty[2]; Huan Huang[3]; and Marc Chioda[3]
[1] US Oncology Network, Blacksburg, VA; [2] Ontada, Woodlands, TX; [3] G1 Therapeutics, Inc., Research Triangle Park, NC

INTRODUCTION

- Myelosuppressive hematological adverse events (HAEs; anemia, neutropenia, and/or thrombocytopenia) are common complications of chemotherapy among patients with cancer[1]
- Cytotoxic chemotherapy remains the cornerstone of treatment for extensive-stage small cell lung cancer (ES-SCLC)[2–4]

OBJECTIVE

- To assess health care resource utilization (HCRU), costs, and treatment patterns associated with myelosuppressive HAEs among patients with ES-SCLC treated with chemotherapy in the community oncology setting

METHODS

Data Source

- This retrospective observational study was conducted using structured data from The US Oncology Network's iKnowMed electronic health record system
- Data on vital status from the Social Security Administration's Limited Access Death Master File and HCRU data from the Financial Data Warehouse were included

Study Population

- Adult patients with ES-SCLC who initiated chemotherapy between January 1, 2015, and December 31, 2019, were stratified into 2 study cohorts on the basis of the presence of grade ≥ 3 HAEs after chemotherapy initiation (index date; **Figure 1**)
 - The cohort with grade ≥ 3 HAEs comprised patients who had ≥ 1 of the following events after index: grade ≥ 3 anemia (hemoglobin <8.0 g/dL), grade ≥ 3 neutropenia (absolute neutrophil count <1000/μL), or grade ≥ 3 thrombocytopenia (platelets <50,000/μL)[5]
 - The cohort without grade ≥ 3 HAEs comprised patients who had no grade ≥ 3 anemia, grade ≥ 3 neutropenia, or grade ≥ 3 thrombocytopenia events after index
 - The first course of chemotherapy initiated after diagnosis of ES-SCLC was defined as chemotherapy initiation; patients must have had no evidence of receiving any chemotherapy within the 12 months prior to diagnosis
- Patients were followed longitudinally from the index date until December 31, 2020, death, or the last patient record, whichever occurred first
- Patients diagnosed with other primary tumors or enrolled in clinical trials during the study period were excluded

Outcome and Analysis

- HAEs were identified using laboratory values from iKnowMed on the basis of Common Terminology Criteria for Adverse Events version 5.0 definitions[5]
- The prevalence and frequency of HAEs (by type and grade), treatment patterns, HCRU (including supportive care utilization [granulocyte colony-stimulating factor {G-CSF}, erythropoiesis-stimulating agents {ESAs}, intravenous {IV} hydration]), and health care costs during the follow-up period were evaluated for both cohorts. Costs were adjusted to the year 2021[6]

Figure 1. Study Design Overview

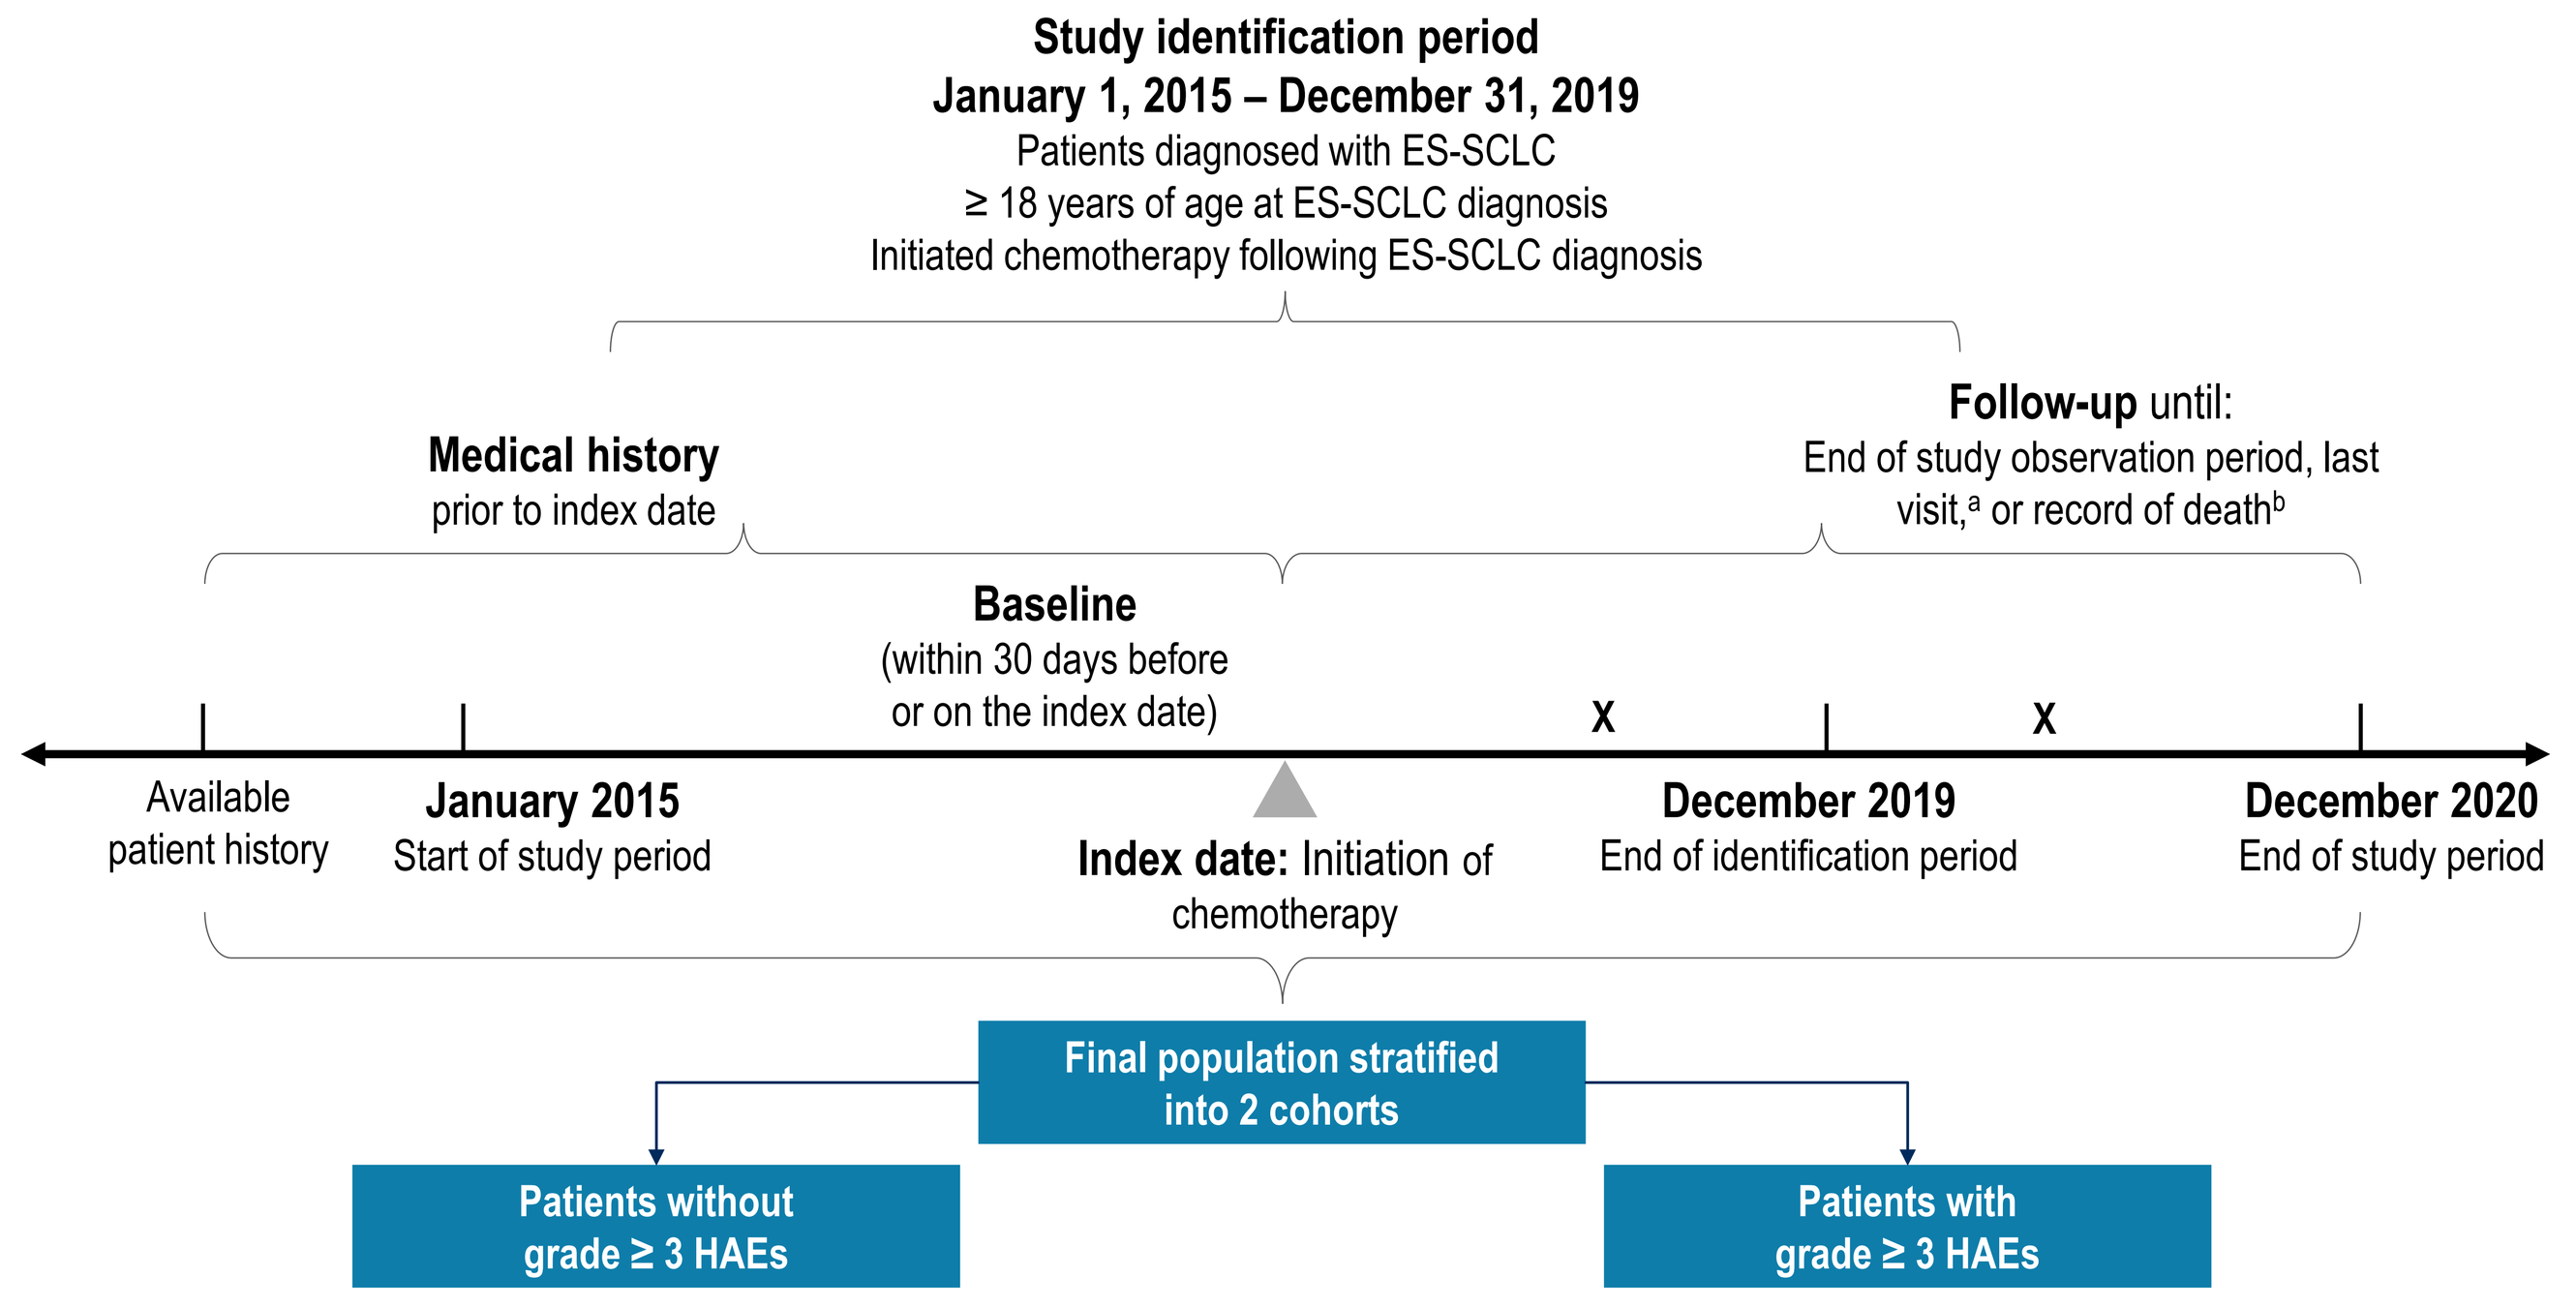
Study identification period
January 1, 2015 – December 31, 2019
Patients diagnosed with ES-SCLC
≥ 18 years of age at ES-SCLC diagnosis
Initiated chemotherapy following ES-SCLC diagnosis
Medical history
prior to index date
Follow-up until:
End of study observation period, last visit,[a] or record of death[b]
Baseline
(within 30 days before or on the index date)
X
X
Available patient history
January 2015
Start of study period
Index date: Initiation of chemotherapy
December 2019
End of identification period
December 2020
End of study period
Final population stratified into 2 cohorts
Patients without grade ≥ 3 HAEs
Patients with grade ≥ 3 HAEs

[a] Last visit = last physical encounter.
[b] Whichever occurred first.
X = additional visit at qualifying USON clinic or record of death; HAE, hematological adverse event; ES-SCLC, extensive-stage small cell lung cancer; USON, US Oncology Network.

RESULTS

Demographic and Clinical Characteristics

- The study population included 778 patients who had ≥ 1 grade ≥ 3 HAE and 596 patients who did not have a grade ≥ 3 HAE after chemotherapy initiation. Demographic and clinical characteristics at baseline are shown in **Table 1**

Myelosuppressive Events

- Among 778 patients in the grade ≥ 3 HAE cohort, 47.3% of patients had grade ≥ 3 anemia, 58.9% had grade ≥ 3 neutropenia, and 37.3% had grade ≥ 3 thrombocytopenia within 12 months post index
 - Mean numbers of events within 12 months post index were 2.0, 1.8, and 2.4 for patients who experienced grade ≥ 3 anemia, grade ≥ 3 neutropenia, and grade ≥ 3 thrombocytopenia, respectively
- 12.2% of patients with grade ≥ 3 HAEs had evidence of major bleeding events (platelets < 20,000/μL)
- Grade ≥ 3 leukopenia and lymphopenia events also occurred in a notable number of patients (**Table 2**)

Treatment Patterns

- Almost all patients (> 99%) received first-line chemotherapy at index (approximately 80% received a platinum-/etoposide-containing regimen and 15% received platinum/etoposide in combination with immunotherapy) in both cohorts
- Patients with grade ≥ 3 HAEs had a higher proportion of dose reductions (46.7% vs 32.2%), treatment holds (12.7% vs 5.9%), and treatment delays between 14–60 days (92.3% vs 84.3%) after chemotherapy initiation (all *P* < 0.001) compared with patients without grade ≥ 3 HAEs (**Table 2**)

Table 1. Demographic and Clinical Characteristics at Baseline

Baseline Characteristic	Cohort With Grade ≥ 3 HAEs (n = 778)	Cohort Without Grade ≥ 3 HAEs (n = 596)	*P*-value
Age group, mean (SD), n	67.2 (9.2)	67.8 (8.9)	0.19
< 65 years	318 (40.9)	239 (40.1)	
≥ 65 years	460 (59.1)	357 (59.9)	
Male sex, n (%)	400 (51.4)	276 (46.3)	0.40
Race, n (%)			0.13
Caucasian	647 (83.2)	489 (82.1)	
African American	40 (5.1)	29 (4.9)	
Asian or other	18 (2.3)	5 (0.8)	
ECOG PS, n (%)			0.16
0	56 (7.2)	52 (8.7)	
1	382 (49.1)	306 (51.3)	
2	158 (20.3)	129 (21.6)	
≥ 3	17 (2.2)	14 (2.4)	
Not documented	165 (21.2)	95 (15.9)	
Count of metastatic site(s) at index, n (%)			< 0.01
1	213 (27.4)	195 (32.7)	
2	110 (14.1)	119 (20.0)	
3	60 (7.7)	61 (10.2)	
≥ 4	49 (6.3)	26 (4.4)	
Not documented	346 (44.5)	195 (32.7)	
Index LOT, n (%)[a]			0.66
LOT 1	776 (99.7)	593 (99.5)	
LOT 2	2 (0.3)	3 (0.5)	
Hemoglobin at baseline, mean (SD), g/dL	12.1 (2.0)	12.5 (1.7)	< 0.01
ANC at baseline, mean (SD), 1000/μL	6.5 (3.5)	6.9 (3.4)	0.06
Platelet count at baseline, mean (SD), 1000/μL	269.4 (111.6)	285.0 (104.4)	0.01
Time from ES-SCLC diagnosis to index date, mean (SD), months	0.9 (4.1)	1.0 (5.1)	0.67

[a] This is the line of therapy for the index regimen received by a patient.
ANC, absolute neutrophil count; ECOG PS, Eastern Cooperative Oncology Group performance status; ES-SCLC, extensive-stage small cell lung cancer; HAE, hematological adverse event; LOT, line of therapy.

HCRU for HAE Management

- 43.1% of patients with grade ≥ 3 HAEs were eligible for RBC transfusions and 3.9% were eligible for platelet transfusions as indicated by laboratory values (**Table 2**)
- Patients with grade ≥ 3 HAEs were more likely to receive therapeutic G-CSF than patients without grade ≥ 3 HAEs (37.8% vs 18.1%; *P* < 0.01), and prophylactic G-CSF use was similar among both cohorts (38.7% vs 49.8%; *P* = 0.40; **Table 2**, **Figure 2**)
 - Receiving G-CSF within 1–3 days after chemotherapy initiation was considered prophylactic use
 - Receiving G-CSF ≥ 4 days after chemotherapy initiation was considered therapeutic use
- The total costs within 12 months post index were higher for patients with grade ≥ 3 HAEs than for those without ($40,896 vs $33,631; *P* < 0.01; **Figure 3**)
- Patients with grade ≥ 3 HAEs had a mean of 10.7 outpatient visits within 12 months post index, versus 7.7 outpatient visits for those without grade ≥ 3 HAEs (*P* < 0.01; **Table 3**)
- Compared with patients without grade ≥ 3 HAEs, patients with grade ≥ 3 HAEs also had greater:
 - G-CSF use (64.1% vs 57.2%; mean number of administrations 3.5 vs 2.4; mean cost per patient $10,943 vs $8821; all *P* < 0.01; **Table 3**, **Figure 3**) within 12 months after the index date
 - ESA use (18.6% vs 4.9%; mean number of administrations 0.7 vs 0.1; mean cost per patient $787 vs $152; all *P* < 0.01; **Table 3**, **Figure 3**) within 12 months after the index date
 - IV hydration use (46.0% vs 30.4%; mean number of administrations 2.3 vs 1.2; mean cost per patient $159 vs $36; all *P* < 0.01; **Table 3**, **Figure 3**) within 12 months after the index date

Table 2. Treatment Outcomes During Follow-up

Outcome During Follow-up	Cohort With Grade ≥ 3 HAEs (n = 778)	Cohort Without Grade ≥ 3 HAEs (n = 596)	*P*-value
Follow-up duration from index date, mean (SD), months	10.5 (8.8)	7.9 (7.9)	< 0.01
Reason for end of follow-up, n (%)			0.09
Death	506 (65.0)	366 (61.4)	
Last activity date on or before study end date	272 (35.0)	230 (38.6)	
Number of chemotherapy cycles, n (%)			< 0.01
1	45 (5.8)	82 (13.8)	
2	68 (8.7)	66 (11.1)	
3	63 (8.1)	52 (8.7)	
4	242 (31.1)	183 (30.7)	
5	64 (8.2)	31 (5.2)	
6	204 (26.2)	142 (23.8)	
> 6	88 (11.3)	37 (6.2)	
Not documented	4 (0.5)	3 (0.5)	
Dose decrease of index treatment, n (%)[a]	356 (46.7)	184 (32.2)	< 0.01
Index treatment hold, n (%)[a,b]	97 (12.7)	34 (5.9)	< 0.01
Index treatment delay, n (%)[a]			
14–60 days	703 (92.3)	482 (84.3)	< 0.01
14–30 days	693 (90.9)	474 (82.9)	< 0.01
31–60 days	175 (23.0)	84 (14.7)	< 0.01
Patients who met transfusion criteria, n (%)			
RBC transfusions (hemoglobin < 8 g/dL)	335 (43.1)	0 (0.0)	< 0.01
Platelet transfusions (platelets < 10,000/μL)	30 (3.9)	0 (0.0)	< 0.01
G-CSF use, n (%)			
Prophylactic	301 (38.7)	297 (49.8)	0.40
Therapeutic	294 (37.8)	108 (18.1)	< 0.01
Type of G-CSF, n (%)			< 0.01
Pegfilgrastim (Neulasta)	531 (68.3)	384 (64.4)	
Filgrastim-sndz (Zarxio)	117 (15.0)	25 (4.2)	
Filgrastim (Neupogen, Accofil)	50 (6.4)	10 (1.7)	
Pegfilgrastim-cbqv (Udenyca)	9 (1.2)	8 (1.3)	
Type of ESA, n (%)			0.96
Darbepoetin alfa (Aranesp)	159 (20.4)	34 (5.7)	
Leukopenia events, n (%)			< 0.01
Grade 3: WBCs 1000–2000/μL	310 (39.8)	33 (5.5)	
Lymphopenia events, n (%)			0.47
Grade 3: Lymphocytes 200–499/μL	328 (42.2)	152 (25.5)	
Grade 4: Lymphocytes < 200/μL	11 (1.4)	4 (0.7)	

[a] Denominator was calculated on the basis of patients with available data and not the full sample population. [b] Treatment hold was defined as a gap of ≥ 60 days without treatment.
ESA, erythropoiesis-stimulating agent; G-CSF, granulocyte colony-stimulating factor; HAE, hematological adverse event; RBC, red blood cell; WBC, white blood cell.

Figure 2. G-CSF Use in Patients With and Without Grade ≥ 3 HAEs

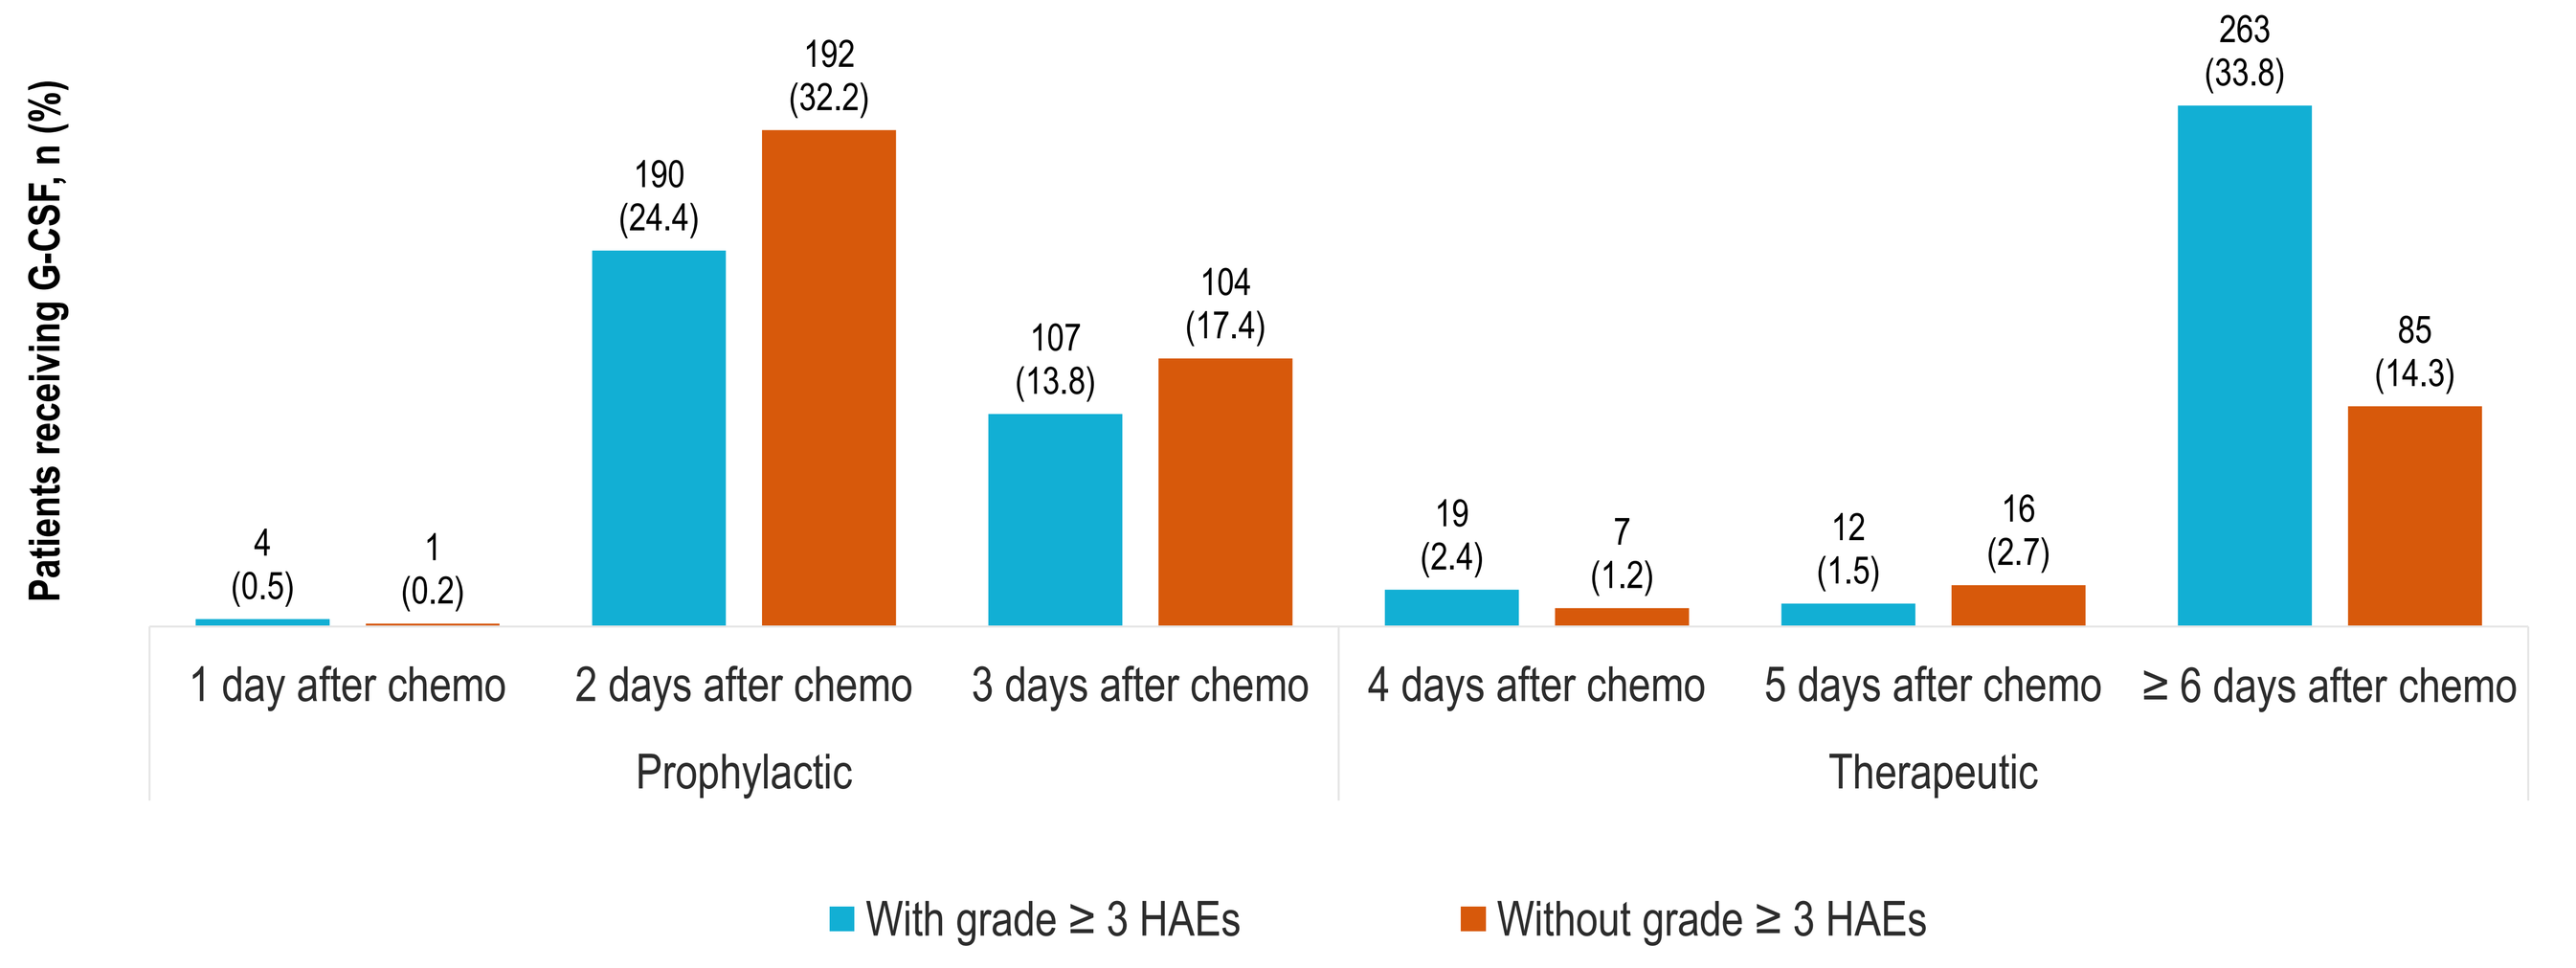
Patients receiving G-CSF, n (%)
4 (0.5)
1 (0.2)
190 (24.4)
192 (32.2)
107 (13.8)
104 (17.4)
19 (2.4)
7 (1.2)
12 (1.5)
16 (2.7)
263 (33.8)
85 (14.3)
1 day after chemo
2 days after chemo
3 days after chemo
4 days after chemo
5 days after chemo
≥ 6 days after chemo
Prophylactic
Therapeutic
With grade ≥ 3 HAEs
Without grade ≥ 3 HAEs

Values above bars indicate patients receiving G-CSF, n (%)
Chemo, chemotherapy; G-CSF, granulocyte colony-stimulating factor; HAE, hematological adverse event.

Table 3. HCRU Within 12 Months After The Index Date – Per Patient

HCRU	Cohort With Grade ≥ 3 HAEs (n = 778)	Cohort Without Grade ≥ 3 HAEs (n = 596)	*P*-value
Total number of outpatient visits, mean (SD)[a]	10.7 (7.9)	7.7 (6.8)	< 0.01
G-CSF use, n (%)	499 (64.1)	341 (57.2)	< 0.01
Number of G-CSF administrations, mean (SD)	3.5 (4.6)	2.4 (3.2)	< 0.01
ESA use, n (%)	145 (18.6)	29 (4.9)	< 0.01
Number of ESA administrations, mean (SD)	0.7 (1.7)	0.1 (0.8)	< 0.01
IV hydration use, n (%)	358 (46.0)	181 (30.4)	< 0.01
Number of IV hydrations, mean (SD)	2.3 (5.5)	1.2 (3.0)	< 0.01

[a] Outpatient visits include physician office visits for new and existing patients, consultations, and follow-up visits. This does not include encounters where a service was rendered (ie, laboratory encounters, minor procedures, etc.).
ESA, erythropoiesis-stimulating agent; G-CSF, granulocyte colony-stimulating factor; HAE, hematological adverse event; HCRU, health care resource utilization; IV intravenous.

Figure 3. Health Care Costs Within 12 months After The Index Date – Per Patient

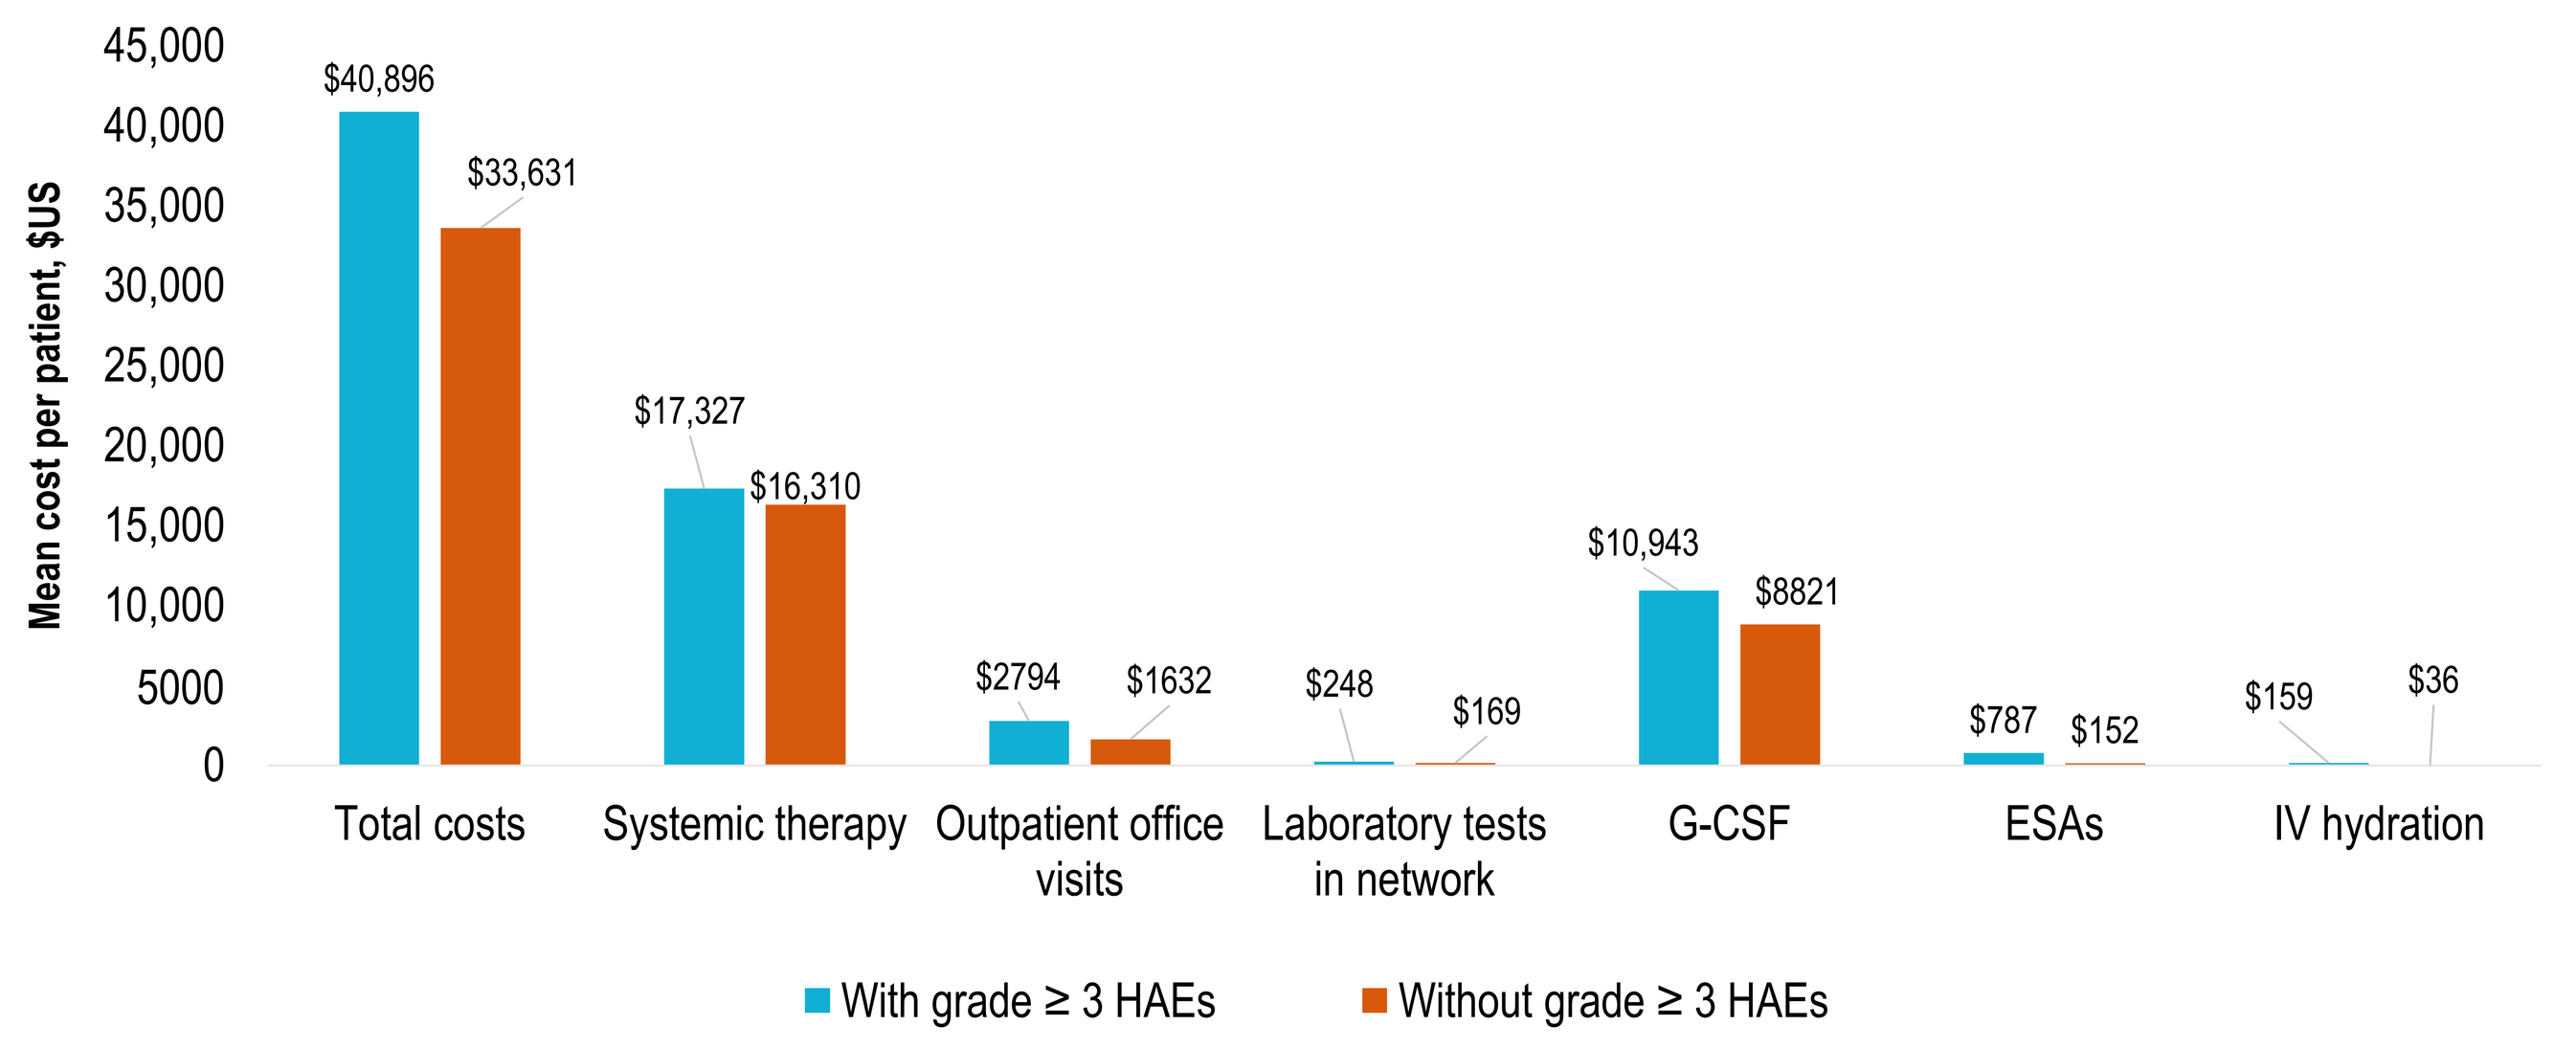
Mean cost per patient, $US
45,000
40,000
35,000
30,000
25,000
20,000
15,000
10,000
5000
0
$40,896
$33,631
$17,327
$16,310
$2794
$1632
$248
$169
$10,943
$8821
$787
$152
$159
$36
Total costs
Systemic therapy
Outpatient office visits
Laboratory tests in network
G-CSF
ESAs
IV hydration
With grade ≥ 3 HAEs
Without grade ≥ 3 HAEs

Only cost categories relevant to the study are highlighted; each category listed reported a *P*-value < 0.01 except systemic therapy costs. All other treatment-related costs were bundled into an "other" category not shown.
ESA, erythropoiesis-stimulating agent; G-CSF, granulocyte colony-stimulating factor; HAE, hematological adverse event; IV, intravenous.

LIMITATIONS

- Results were based on data from community oncology settings and may not be generalizable beyond this setting
- Data in the inpatient settings were not captured; inpatient costs or costs for transfusions were not included

CONCLUSIONS

- The results suggest there is significant burden of myelosuppressive HAEs on patients with ES-SCLC in a community oncology setting
- Patients with grade ≥ 3 HAEs had more dose reductions, treatment delays, and HCRU than those without grade ≥ 3 HAEs
- Therapies to protect bone marrow from multilineage HAEs have the potential to reduce such burden. Future research should investigate HCRU and cost burden in the inpatient setting to better understand the full scope of HAE management

ASH Annual Meeting | December 11–14, 2021

REFERENCES:
1. Kurtin S. *J Adv Pract Oncol.* 2012;3:209–24.
2. Aapro M, et al; ESMO Guidelines Committee. *Ann Oncol.* 2018;29(suppl 4):iv96–110.
3. Crawford J, et al. *Support Care Cancer.* 2020;28:925–32.
4. Kuter DJ. *Oncology (Williston Park).* 2015;29:282–94.
5. National Cancer Institute. Common Terminology Criteria for Adverse Events (CTCAE) Version 5.0. US Department of Health and Human Services: NIH. https://ctep.cancer.gov/protocoldevelopment/electronic_applications/docs/CTCAE_v5_Quick_Reference_8.5x11.pdf. Accessed September 22, 2021.
6. Bureau of Labor Statistics. Medical Care Consumer Price Index. https://www.bls.gov/cpi/factsheets/. Accessed October 7, 2021.

ACKNOWLEDGMENTS:
- Study sponsored by G1 Therapeutics.

DISCLAIMER:
This presentation is the intellectual property of the author/presenter. Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors. Contact them at hhuan@g1therapeutics.com for permission to reprint and/or distribute.